Time-assessed infra-red thermal characterization of canine cutaneous mast cell tumors (cMCT) treated intratumorally with the investigational anticancer agent Tigilanol Tiglate (EBC-46)

MELO[1], Samanta Rios; JANUÁRIO[1], Eric Vieira; ZANUTTO[1], Erika; LOWDEN[2], Stewart; MATERA[1], Julia Maria

1- Department of Surgery - School of Veterinary Medicine and Animal Science – University of São Paulo, Brazil
2- QBiotics Group - Queensland - Australia

INTRODUCTION

Tigilanol Tiglate Injection is a product derived from the seed of native Australian blushwood tree (*Fontainea picrosperma).* It has been delivered intratumorally to over 250 cMCT. Thermographic imaging (TI) was previously described in evaluating temperature differences between tumor and healthy skin of 78 cMCT[1], providing key information about tumor vascular changes. This study is investigating time-assessed TI of cMCT treated with tigilanol tiglate (EBC-46).

MATERIALS AND METHODS

TI of a range of clinically presented cMCT were collected at T0 (pre-treatment); T1 (during Tigilanol Tiglate Injection); 2h, 4h and at 1, 7, 14, 28 days post injection. Tumors were also CT scanned prior to treatment and at Day 28.

RESULTS

Evidence is presented showing initial approximate 4ºC decrease in Tumoral Temperature (TT) followed by measurable TT increase during subsequent hours (1-2ºC). In subsequent days, TT again decreases and remains lower (around 5ºC) than prior to treatment. By Day 28, the resolving injection site resembles the temperature in surrounding healthy tissue. Tumor measurements were also collected and RECIST results also presented. The drug was well tolerated with no adverse effects of treatment, no tumor recurrence.

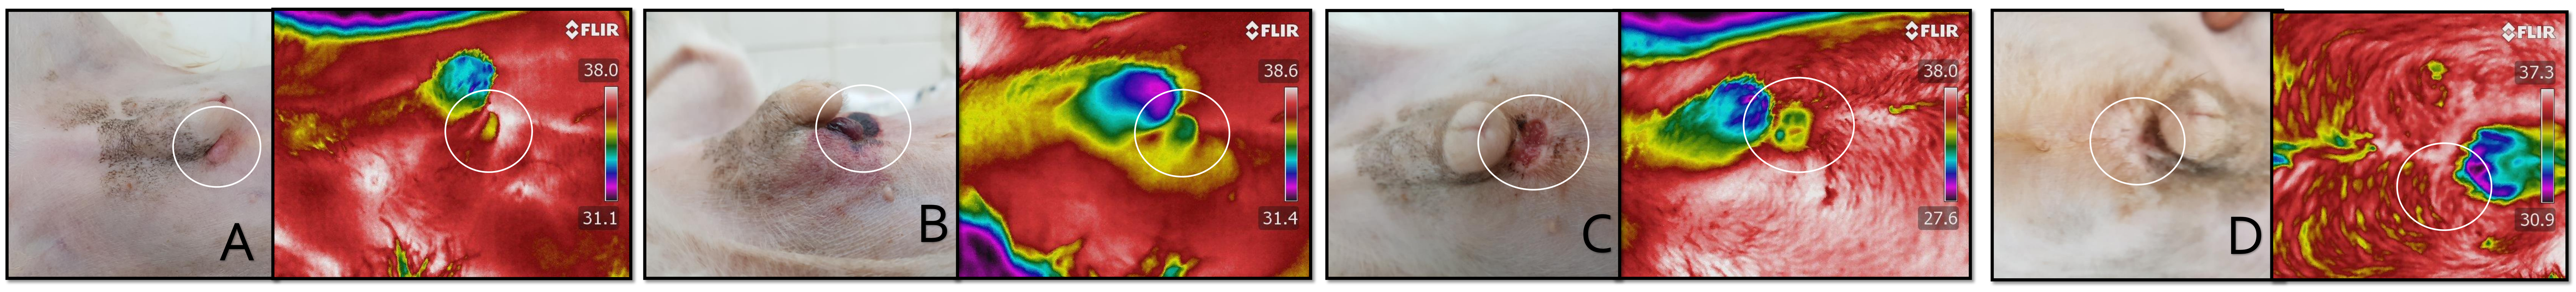
A
FLIR
38.0
31.1
B
FLIR
38.6
31.4
C
FLIR
38.0
27.6
D
FLIR
37.3
30.9

Figure 1: *Evolution of mast cell tumor treated with Tigilanol Tiglate Injection – Animal number 1 – (A) Before injection on Day 0 – white circle indicates MCT; (B) Day 1; (C) Day 4; (D) Day 28, after RECIST complete response of tumor. (left images are white light, and right are thermographic images).*

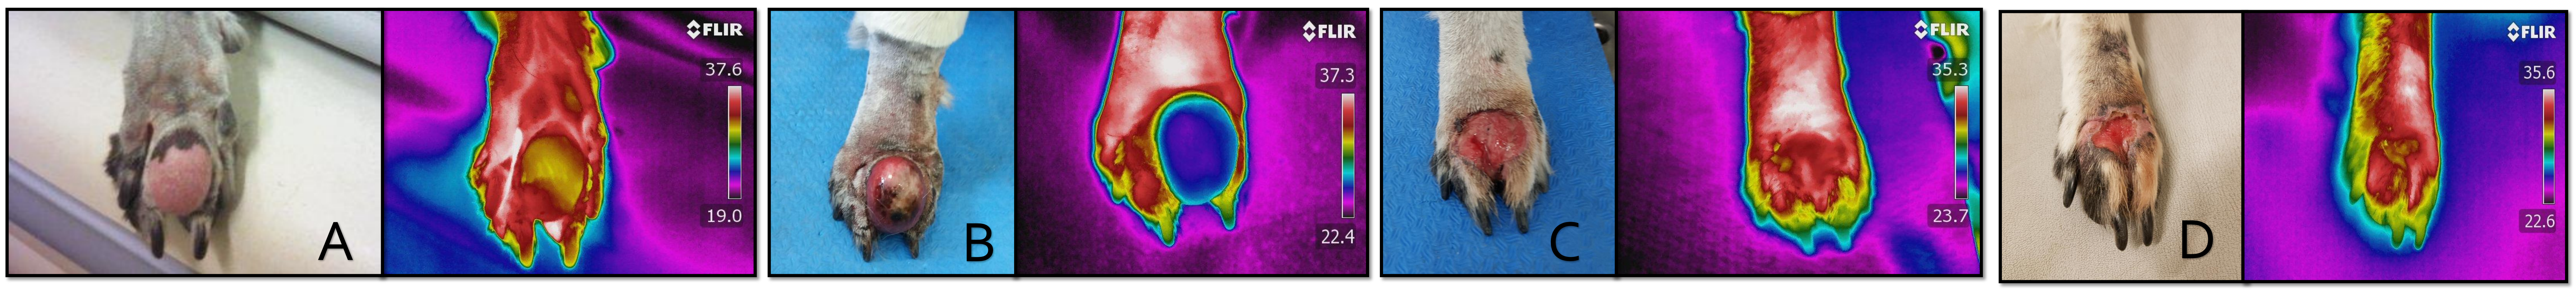
A
FLIR
37.6
19.0
B
FLIR
37.3
22.4
C
FLIR
35.3
23.7
D
FLIR
35.6
22.6

Figure 2: *Evolution of mast cell tumors treated with Tigilanol Tiglate Injection – Animal number 5 – (A) Before injection Day 0; (B) Day 1; (C) Day 14; (D) Day 28, after RECIST complete response of tumor. (left images are white light, and right are thermographic images).*

CONCLUSION

Tigilanol tiglate treatment results in vascular disruption and an acute highly-localized inflammatory response at injection site. During subsequent days, TT remains lower (-5ºC) than prior to treatment, explained by drug-induced tumoral necrosis. Injection site temperatures return similar to those of surrounding skin at Day 28, with resultant tumor removal and rapid wound resolution. Tigilanol Tiglate Injection is currently being evaluated under an Investigational New Animal Drug with FDA-CVM in the US where Pivotal Field Efficacy study treating cMCT is underway.

REFERENCES

Melo, S.R., Macedo, T.R., Cogliati, B., Ferrigno, C.R.A., Matera, J.M. (2015) Thermographic assessment of canine mast cell tumours. *Indian Journal of Applied Research* 5(3): 539-543.

Veterinary Cooperative Oncology Group – common terminology criteria for adverse events (VCOG-CTCAE) following chemotherapy or biological antineoplastic therapy in dogs and cats v1.1, *Vet Comp Oncol.* July 2011. Available from: < http://www.vetcancersociety.org/members/files/2012/01/CTCAE-v1-1.pdf> [18 Mar 2015].

Monga, S.P., Wadleigh, R., Sharma, A., Adib, H., Strader, D., Singh, G., Harmon, J.W., Berlin, M., Monga, D.K. and Mishra, L. (2000) Intratumoural therapy of cisplatin/epinephrine injectable gel for palliation in patients with obstructive esophageal cancer. *Am J Clin Oncol (CCT)* 23: 386-392.